A Phase 2 Study of Peginterferon Lambda, Lonafarnib, and Ritonavir for 24 Weeks: End-of-Treatment Results from the LIFT HDV Study.

Christopher Koh[1], Julian Hercun[1], Farial Rahman[1], Amy Wen-Chun Huang[1], Ben L. Da[1], Pallavi Surana[1], Devika Kapuria[1], Yaron Rotman[1], Anusha Vittal[1], Christy Ann Gilman[1], Gil Ben-Yakov[1], Walter Chunwei Lai[1], Harel Dahari[2], Jeffrey S. Glenn[3], and Theo Heller[1]

National Institute of Diabetes & Digestive & Kidney Diseases[1], Loyola University Medical Center[2], Stanford University School of Medicine[3]

Introduction

- 15-20 million people are infected worldwide with chronic hepatitis D (HDV).
- Up to 80% of patients with HDV may develop cirrhosis within 5-10 years.
- Interferon-alfa based therapies are unsatisfactory, <30% achieve HBsAg loss and become HDV RNA negative.
- Separately, peginterferon lambda (LMD) and the prenylation inhibitor lonafarnib (LNF) +/- ritonavir (RTV) boosting have demonstrated effectiveness against HDV in early phase clinical trials.

Aims

- To assess the antiviral effects and safety of combination therapy with Peginterferon Lambda, Lonafarnib and Ritonavir in patients with chronic HDV infection.

Methods

- In this phase 2a open-label study, 26 adult patients chronically infected with HDV and quantifiable HDV RNA in serum were dosed for 24 weeks with:
 - Peginterferon lambda (LMD) 180 mcg subcutaneously weekly
 - Lonafarnib (LNF) 50 mg orally twice daily
 - Ritonavir (RTV) 100 mg orally twice daily
- Patients were on hepatitis B nucleos(t)ide analogue therapy for at least 3 months with HBV DNA <21 IU/mL prior to initiating therapy.
- Serial measurements of safety parameters, liver tests, virologic (HDV RNA & HBV DNA) and pharmacokinetic markers and symptom questionnaires were performed.
- After completing therapy, patients were followed for an additional 24 weeks.

Results

Study Flow Diagram

Enrolled (n=31)

Excluded (n=2)
Withdrew consent - 2

Screened for Eligibility (n=29)

Excluded (n=3)
Failure to complete screening - 2
Unwilling to undergo liver biopsy - 1

Treatment x 24 weeks (n=26)

Post-Therapy Follow-up x 24 weeks

Baseline Demographics

Characteristic/Lab	Result (n=26)
Male Sex (%)	17 (65)
Median Age (IQR)	40 (36, 46)
Caucasian, Black, Asian (%)	8 (31), 4 (15), 14 (54)
ALT U/L	64 (45, 93)
AST U/L	47 (38, 64)
HBV DNA IU/mL	<20 (<20, 20)
HDV RNA log IU/mL	4.74 (4.18, 5.65)
Median VCTE kPa	11.8 (6, 15)
Ishak Fibrosis	3 (2-4)

Treatment Response at 12 & 24 Weeks

Treatment Duration	Mean Change in log HDV RNA (IU/mL)	Confidence Interval
12 Weeks	3.36	(2.86-3.85)
24 Weeks	3.21	(2.50-3.93)

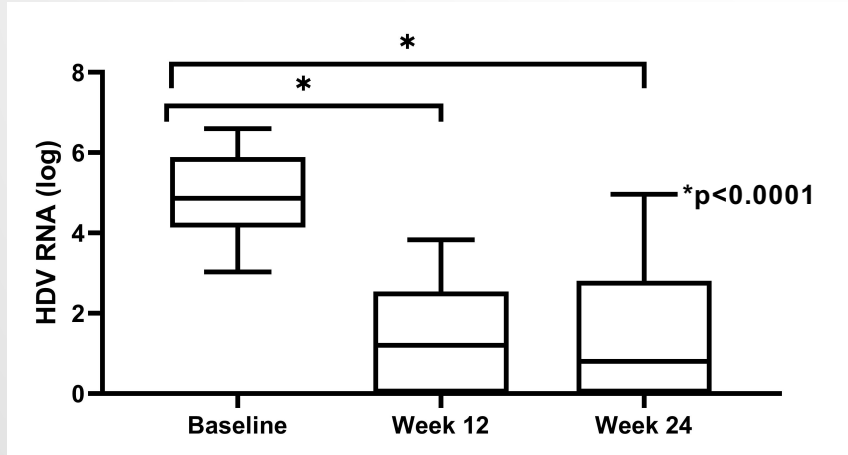
HDV RNA (log)
0
2
4
6
8
*
*
*p<0.0001
Baseline
Week 12
Week 24

HDV RNA Change from Baseline to End of Therapy

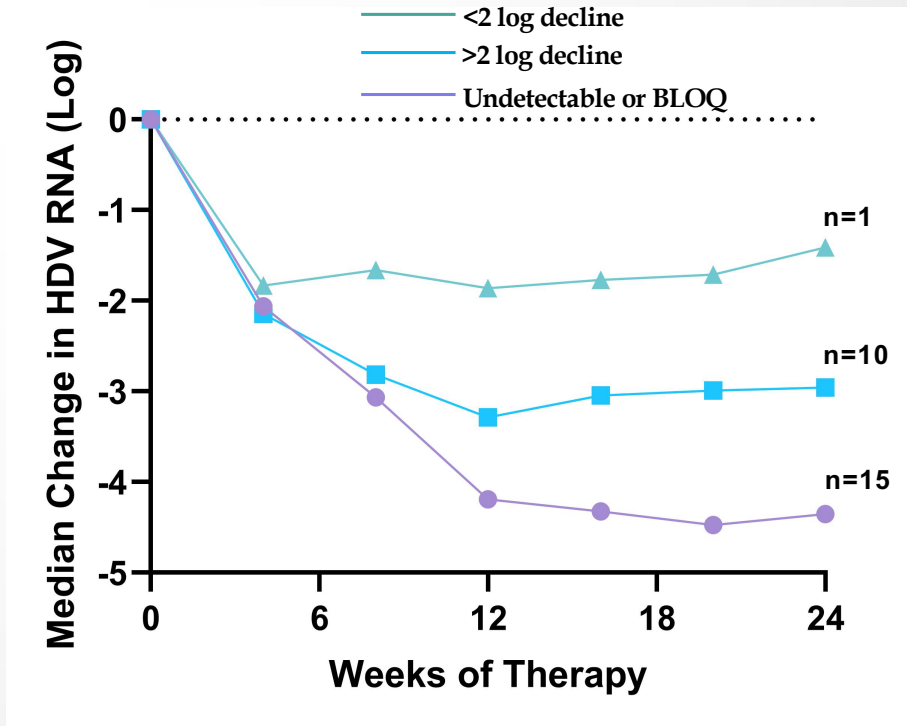
<2 log decline
>2 log decline
Undetectable or BLOQ
Median Change in HDV RNA (Log)
0
-1
-2
-3
-4
-5
n=1
n=10
n=15
0
6
12
18
24
Weeks of Therapy

Adverse Events

Most Common Adverse Events								
Nausea	Diarrhea	Anorexia	Abdominal Bloating	GERD	Fatigue	Weight Loss	Anemia	Hyperbilirubinemia
69%	100%	46%	63%	65%	42%	31%	23%	19%

Dose Reductions/Discontinuations			
	Hyperbilirubinemia	Anemia	Ascites
Dose Reduction	1	1	
Discontinuation	4		1

Discussion

- Compared to baseline, significant declines in HDV RNA levels were demonstrated at both 12 and 24 weeks of therapy with LMD/LNF/RTV.
- 25 of 26 (96%) patients achieved >2 log decline during 24 weeks of therapy with LMD/LNF/RTV.
- By the end of therapy, 58% of subjects achieved BLOQ or undetectable HDV RNA levels.
- Therapy with LMD/LNF/RTV was relatively safe in most patients for up to 6 months.
- Most of the GI related side effects were grade 1 in nature and subsided by week 4 of therapy.
- Per protocol discontinuation of triple therapy was mostly due to known side effects related to peginterferon lambda.
- Currently, all patients in this study have completed therapy and are in various phases of post-treatment follow-up.

Conclusion

- After 24 weeks of therapy, almost all subjects achieve >2 log decline in HDV RNA with 58% achieving undetectable or BLOQ HDV RNA levels.
- These end-of-treatment results support continued exploration of this therapeutic combination in HDV.

References

1. Koh C, et al. Gastroenterol 2019; 156
2. Colombo M, et al. Gastroenterol 1983;85
3. Koh C, et al. Lancet Infect Dis 2015;15
4. Yurdaydin, et al. Hepatology 2018; 67: 1224-1236
5. Wedemeyer, et al. J Hepatol 2018; 66: S24
6. Etzion, et al. J Hepatol 2019; 70, e32

Disclosures

- This work was supported by the intramural research program of the NIDDK NIH.
- All patients provided informed consent for IRB approved studies.
- H. Dahari: Eiger BioPharmaceuticals, NIH RO1AI146917
- J. Glenn: Eiger BioPharmaceuticals, Riboscience, LLC
- All other authors have no financial disclosures.